Forced Degradation Automation

Authors: Lina Liu[1*], Tim Rhodes[1], Roy Helmy[1], Elise Miller[1], Jia Zang[1], Margaret Figus[1], Wes Schafer[1], Paul Harmon[1], Brian Farrer[1], Carlos Perez[2], Scott Donenfeld[2]

Affiliations: [1] – Merck & Co., Inc; [2] – Leap Technologies

Abstract

Forced degradation is an important method used in drug development in the pharmaceutical industry. Forced degradation studies are used for multiple purposes, including demonstration of the specificity of separation methods, gaining insight into degradation pathways, and discernment of degradation products in formulations that are related to drug substances versus those that are related to other ingredients of a formulation.

However, FDA guidance for forced degradation is vague with respect to experimental conditions. In order to harmonize the procedures of forced degradation, an automated method for forced degradation was developed, utilizing the CTC LEAP PAL workstation automation system.

The Automated Forced Degradation approach significantly reduces the amount of manual labor used to perform the tests and harmonizes the operational procedures of forced degradation.

The Automated Forced Degradation system is user-friendly and is intended to be used as a "walk-up system" that is able to prepare forced degradation and linearity samples, perform on-line HPLC analysis as well as generate reports automatically. The details of the system will be discussed along with a number of case studies demonstrating its use.

Conditions and Case Study

Automated Forced Degradation Conditions

Stress Type	*Major Degradation Pathway*	*Reagents*	*Co-solvent (if necessary)*	*API Conc.*	*Temp.*	*Sampling Time*	*Comments*
Primary Solution Phase Conditions							
Acid/Base	**Hydrolysis**	**0.01N HCl/ 0.01N NaOH**	**Acetonitrile**	**Analysis concentration***	**40° C (increase if needed)**	**3 hours**	**Alcohol solvents can facilitate acid/base catalyzed addition of alcohol**
Acid/Base	**Hydrolysis**	**0.1N HCl/ 0.1N NaOH**	**Acetonitrile**	**Analysis concentration***	**40° C (increase if needed)**	**24 hours**	**Alcohol solvents can facilitate acid/base catalyzed addition of alcohol**
H_2O_2	**Nucleophilic Oxidation**	**0.3 % H_2O_2/pH 7 buffer solution**	**MeOH or EtOH**	**Analysis concentration***	**RM**	**24 hours**	**Acetonitrile solvent react with H2O2 to form peroxycarbamic acid**
AIBN	**Auto-oxidation**	**5 mM AIBN in MeOH/ H_2O (1:1)**	**See comments**	**Analysis concentration***	**40° C**	**24 hours**	**Severe solvent effects currently under investigation**
Thermal	**Heat**	**NA**	**Acetonitrile**	**Analysis concentration***	**100° C**	**4 hours**	

User Friendly "Walk Up System"

Sample Preparation only – Cycle Composer

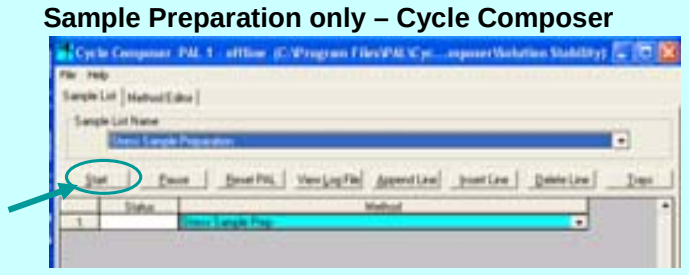

Online sample preparation and analysis –ChemStation/Altas

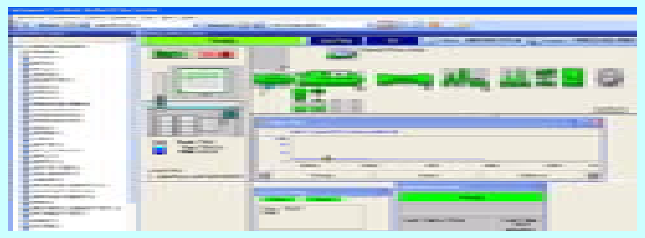

Conclusion and Future Work

Conclusions

- **The LEAP system has been approved and validated for use in performing method validations and forced degradation studies**
- **Linearity, reproducibility, ease of use, and open access have been demonstrated**
- **The automated sample preparation, online analysis, and auto report streamline pre- PCC chemical stability screen**

How Does the Leap Work?

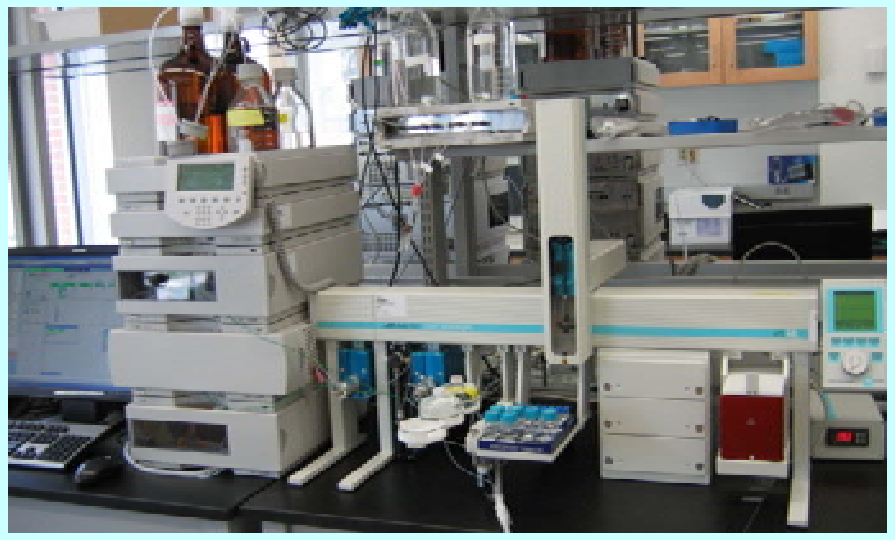

Establishing Linearity Manual vs. LEAP

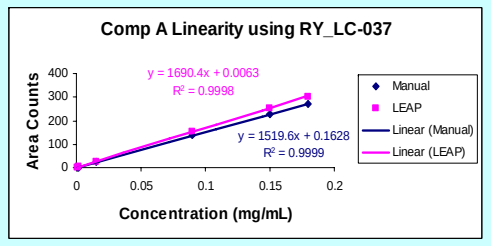
Comp A Linearity using RY_LC-037
y = 1690.4x + 0.0063
R² = 0.9998
y = 1519.6x + 0.1628
R² = 0.9999
Area Counts
Concentration (mg/mL)
Manual
LEAP
Linear (Manual)
Linear (LEAP)

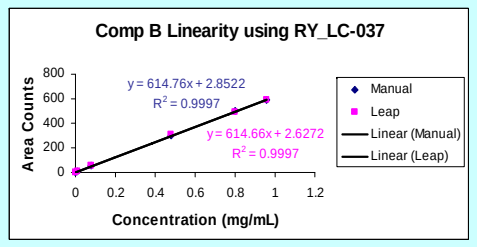
Comp B Linearity using RY_LC-037
y = 614.76x + 2.8522
R² = 0.9997
y = 614.66x + 2.6272
R² = 0.9997
Area Counts
Concentration (mg/mL)
Manual
Leap
Linear (Manual)
Linear (Leap)

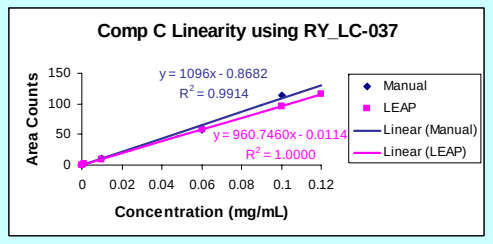
Comp C Linearity using RY_LC-037
y = 1096x - 0.8682
R² = 0.9914
y = 960.7460x - 0.0114
R² = 1.0000
Area Counts
Concentration (mg/mL)
Manual
LEAP
Linear (Manual)
Linear (LEAP)

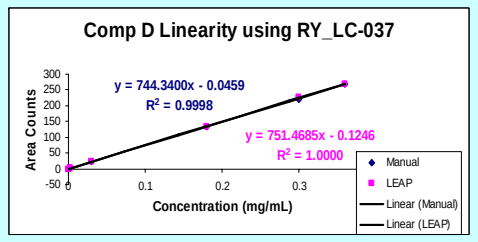
Comp D Linearity using RY_LC-037
y = 744.3400x - 0.0459
R² = 0.9998
y = 751.4685x - 0.1246
R² = 1.0000
Area Counts
Concentration (mg/mL)
Manual
LEAP
Linear (Manual)
Linear (LEAP)

DART for Quick ID of Degradants?

Direct Analysis in Real Time (DART) technology to interface with existing LC/MS

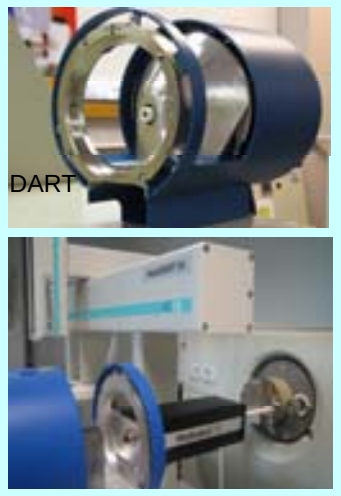
DART

- **Gas-phase proton transfer reactions**

$He^* + nH_2O \longrightarrow He + [(H_2O)_{n-1}H]^+ + OH^-$

$[(H_2O)_{n-1}H]^+ + AB \longrightarrow ABH^+ + (n-1)(H_2O)$

- **Gas-Phase Chemical Ionization**

$[(H_2O)_{n-1}H]^+ + NH_3 \longrightarrow NH_4^+ + (n-1)(H_2O)$

$NH_4^+ + AB \longrightarrow [AB + NH_4]^+$

Future Work

- **Develop and validate forced degradation workflow for lead Op activities**
- **Demonstrate the use of the LEAP System as a method development tool for kinetic profiling, on-line MS ID, etc.**
- **Evaluate and develop low throughput solubility/stability/redispersability combined workflow**
- **Evaluate low throughput dissolution screen**

Goals of the Automated System

- User friendly format – a "Walk-Up System"
- Reproducibly and accurately performs method validation and forced degradation
- Automated Report featuring linearity, LOQ, LOD, and degradation profile overlays
- Adaptability of system to changing conditions (solvents, degradation conditions, etc.)

Automated Report Stress Degradation Overlays

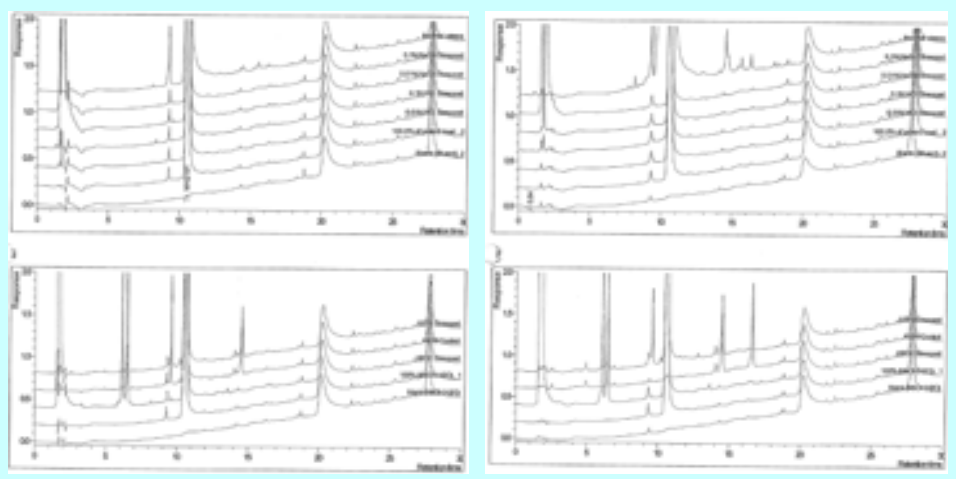

Comp C Manual Preparation Comp C LEAP Preparation

Dart Analysis Compound E

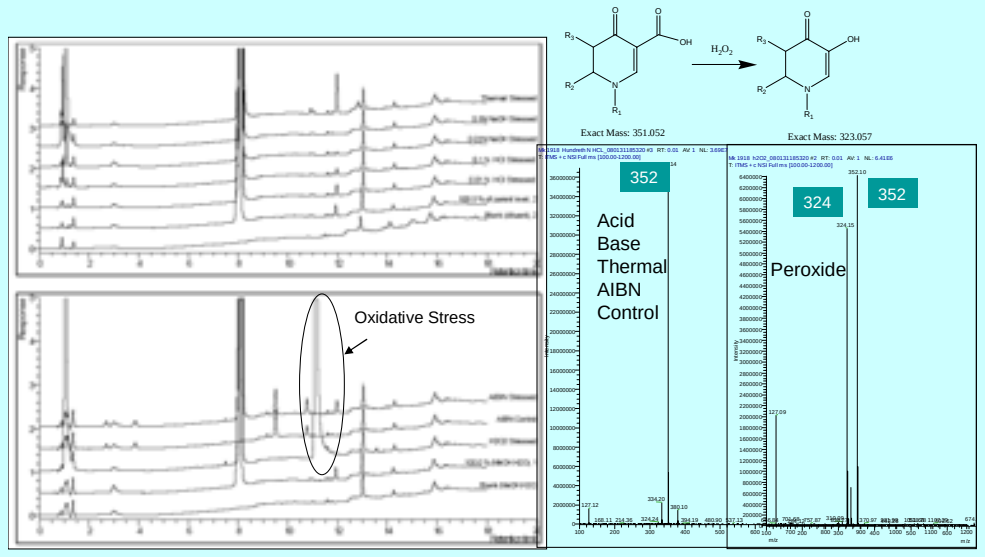
Oxidative Stress
H_2O_2
Exact Mass: 351.052
Exact Mass: 323.057
352
Acid
Base
Thermal
AIBN
Control
324
352
Peroxide

Acknowledgements

Merck

- **Lou Crocker, Mike Riebe, Alexander Chin**
- **Zhihong Ge, Chris Welch, Vinni Antonucci, Bing Mao, Abrahim Ahmed**
- **Xioayi Gong, Adam Beard, Biba Mirlinda, Naijun Wu**
- **Allen Templeton, Andrey Peresypkin**
- **John Delello, JJ Yates, Neil Rissen, Donglan Cai, Doug West, Adam Duckworth**

Leap Technologies

- **Carlos Perez, Scott Donenfeld, Brian Peat**

IonSense

- **Brian Musselman**

Restricted
Confidential
limited access